Budget Savings from Patent Expiration of Cancer Drugs in South Korea

Lee SM[1], Suh D[2], Spiteri C[3], Joung KI[4], KIM ES[5], Suh DC[4]

[1]Daegu Catholic University, Gyeongsan, Korea, [2]Michigan University, Ann Arbor, MI, USA, [3]MSD, Macquarie Park, NSW, Australia, [4]Chung-Ang University, Seoul, Korea, [5]MSD Korea, Seoul, Korea

BACKGROUND

- ❑ Oncology drugs account for the largest segment in any specialty medical area with rapidly growing expenditures, which has been recognized as a major concern worldwide.
- ❑ The introduction of generics after the loss of patent exclusivity plays a major role in budget savings by significantly decreasing drug prices. The loss of exclusivity of originators provides significant savings for patients, healthcare payers, and the government and thus increases the affordability of innovative drugs and expands the coverage of existing drugs.

OBJECTIVES

- ❑ To estimate budget savings from using off-patented oncology medicines, and to inform decision makers how these savings could be used to fund improvement of affordability to new innovative cancer treatments in South Korea.

METHODS

Model structure

- ❑ A model was developed to calculate budget savings due to the patent expiration of cancer drugs in a 5-year period from 2020 to 2024.
- ❑ It was operationalized with market dynamic variables including market share of generics, market growth of therapeutic classes in which the originator will lose market exclusivity, price level of the originator, and price level of generics after patent expiration.

Study drug selection

- ❑ This study selected oncology medicines with one or more valid product patents that will expire during 2020-2024 in Korea.

- Group A (i.e., base case group): at least one valid patent that would expire during the study period.
- Group B (i.e., delayed group): at least one patent was expired prior to the beginning of the study period, but later patents would expire during the study period.
- Group C (i.e., opportunistic group): drugs classified in group A and group B and drugs that were off-patent as of 31 December 2019; however, the exact date of patent expiration was unavailable.

Figure 1. Study cancer drugs included in the analyses

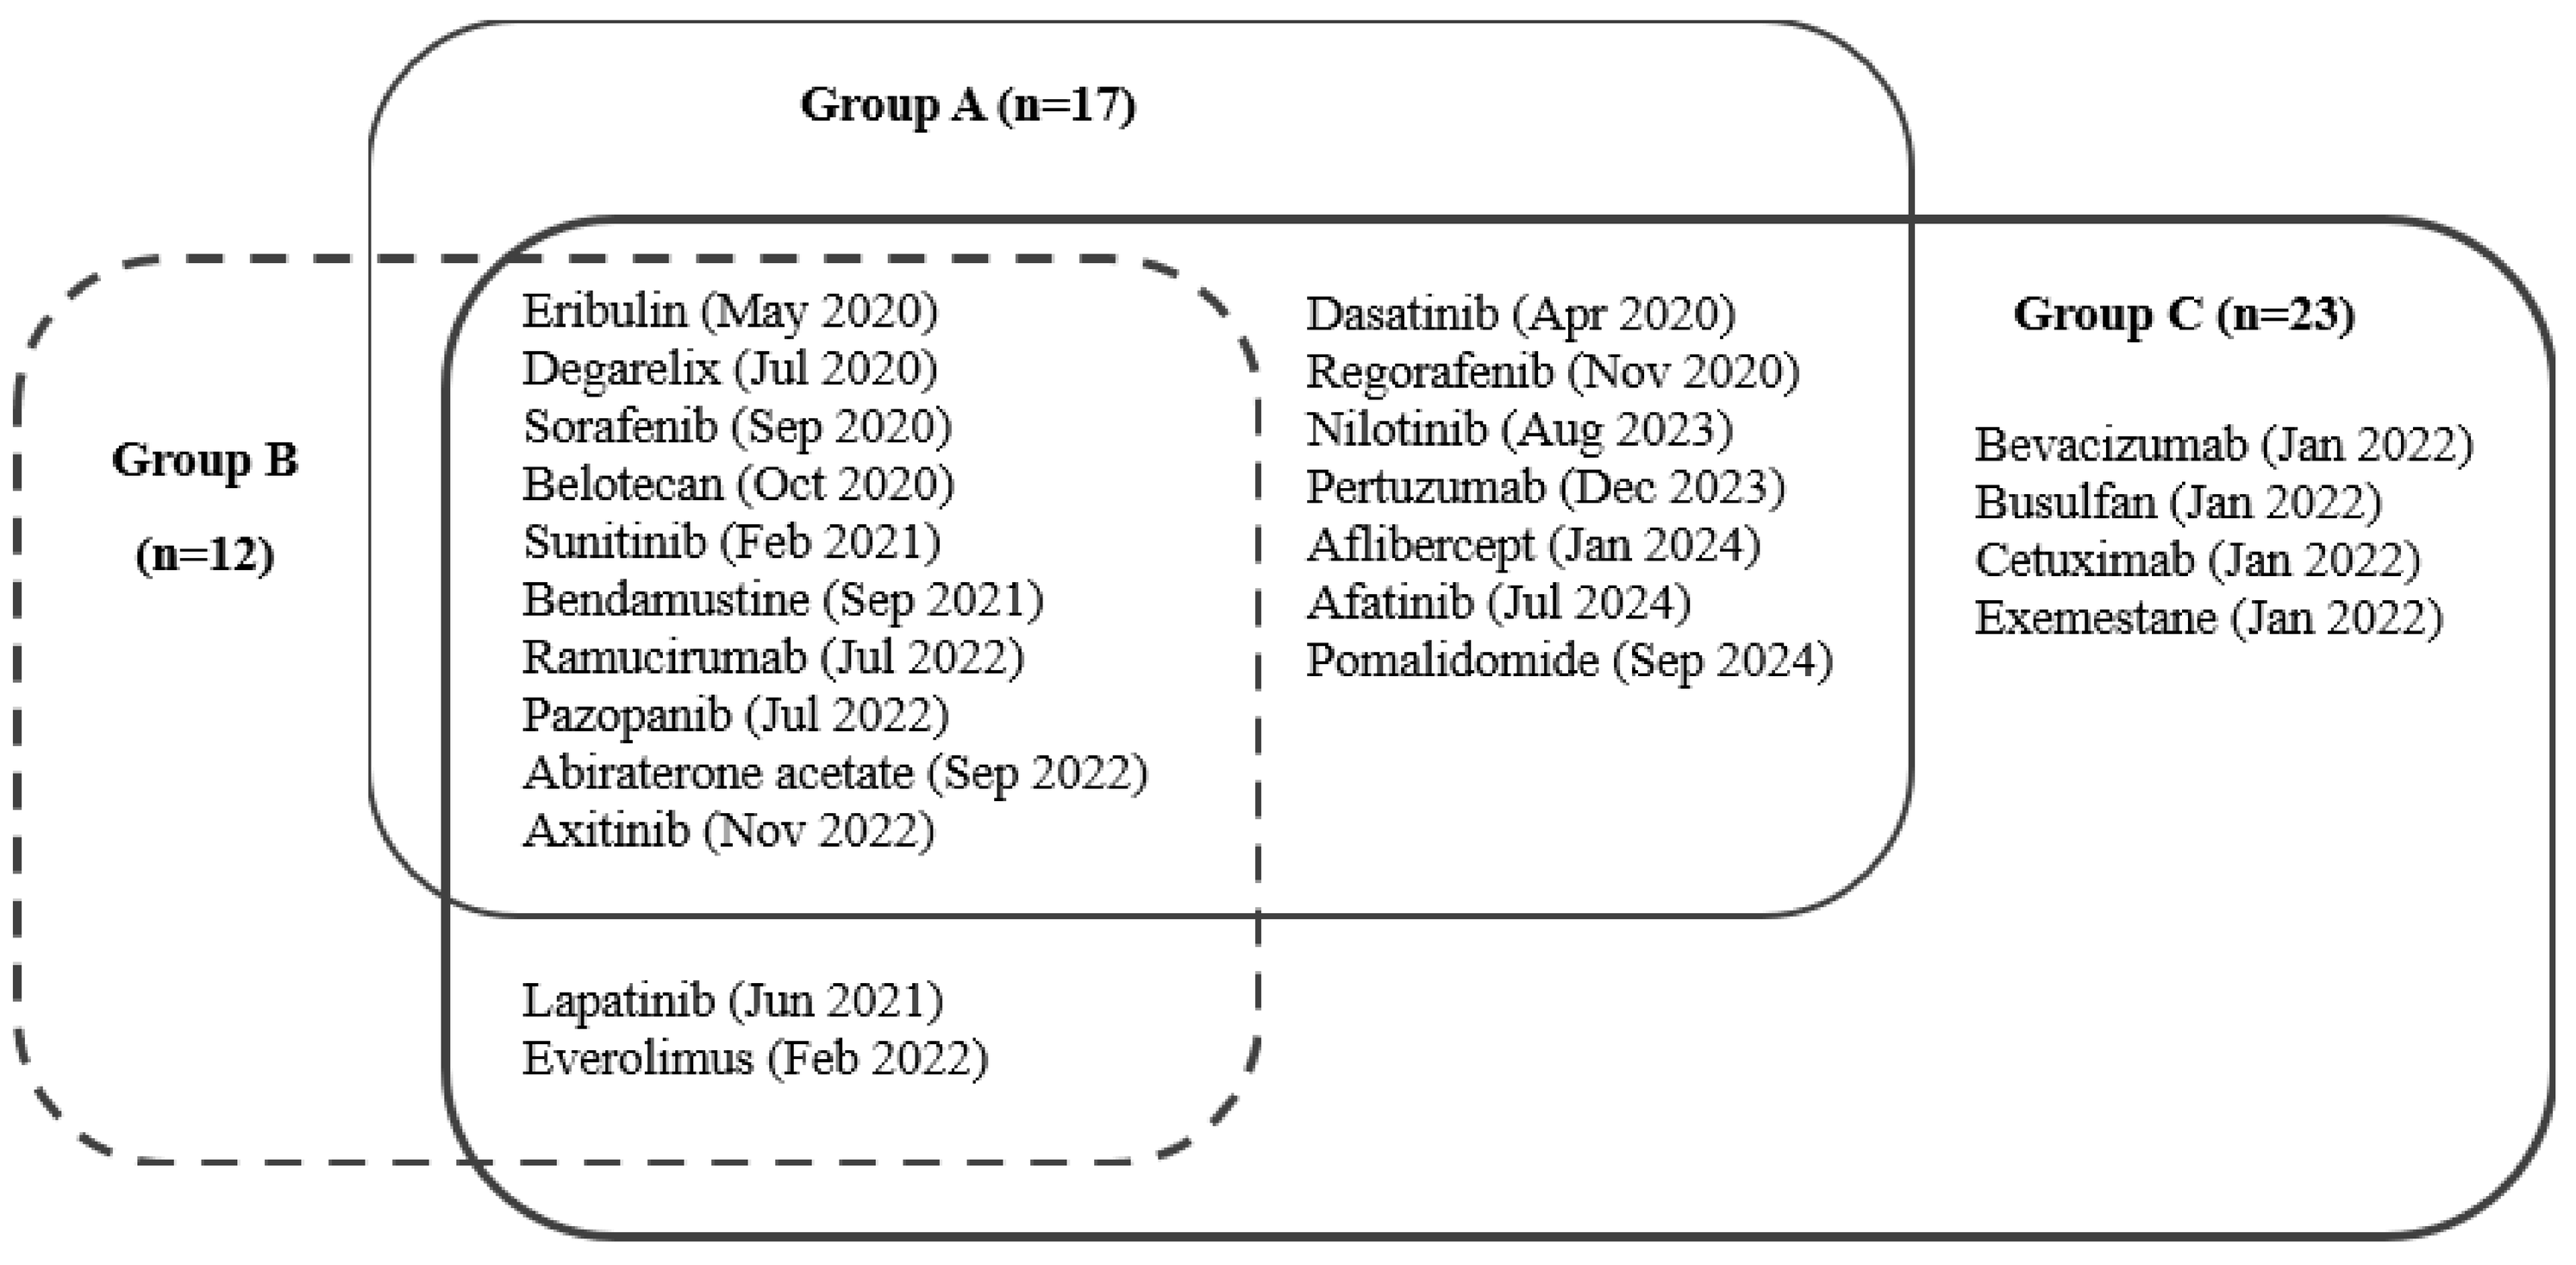
Group A (n=17)
Group B
(n=12)
Group C (n=23)
Eribulin (May 2020)
Degarelix (Jul 2020)
Sorafenib (Sep 2020)
Belotecan (Oct 2020)
Sunitinib (Feb 2021)
Bendamustine (Sep 2021)
Ramucirumab (Jul 2022)
Pazopanib (Jul 2022)
Abiraterone acetate (Sep 2022)
Axitinib (Nov 2022)
Dasatinib (Apr 2020)
Regorafenib (Nov 2020)
Nilotinib (Aug 2023)
Pertuzumab (Dec 2023)
Aflibercept (Jan 2024)
Afatinib (Jul 2024)
Pomalidomide (Sep 2024)
Bevacizumab (Jan 2022)
Busulfan (Jan 2022)
Cetuximab (Jan 2022)
Exemestane (Jan 2022)
Lapatinib (Jun 2021)
Everolimus (Feb 2022)

Month and year of patent expiration are listed in the parenthesis

Analysis of proxy cases

- ❑ An analysis of proxy cases was conducted to improve the accuracy of dynamic market variables during the study period based on the historical trends of the off-patent market for 2012–2018.

Table 1. Input parameters for market dynamics after patent expiration

Variables	Years after patent expiration				
	1st year	2nd year	3rd year	4th year	5th year
Market share of generics/biosimilar					
Chemical generics	5.0%	7.1%	9.2%	11.3%	13.4%
Biosimilars	3.1%	12.5%	12.5%	12.5%	12.5%
Market growth					
Chemical drugs	1.3%	1.4%	1.4%	1.4%	1.4%
Biologics	4.0%	0.0%	0.0%	0.0%	0.0%
Price level of originators*					
Chemical drugs	75%	60%	60%	53%	53%
Biologics	80%	80%	80%	80%	80%
Price level of generics/biosimilars*					
Chemical generics	58%	57%	55%	51%	51%
Biosimilars	72%	57%	57%	57%	57%

* For the price level, the percentages at each year after patent expiration are the relative ratio compared with the price (=100%) at the time of patent expiration.

Sensitivity analyses

- ❑ One-way sensitivity analyses were performed to measure the relative impact of key input parameters on the robustness of the base case budget savings.

RESULTS

Table 2. Annual drug expenditures and budget savings from patent expiration during 2020–2024

Year	Expected drug expenditures		Budget savings*		
	Korea (₩): million	US ($): million	Korea (₩): million	US ($): million	(% of 2019 expenditure)
2019	₩229,094	$198			
2020	₩220,464	$191	₩8,631	$7	4%
2021	₩196,173	$170	₩33,109	$29	14%
2022	₩180,946	$157	₩48,149	$42	21%
2023	₩169,595	$147	₩59,499	$51	26%
2024	₩144,054	$125	₩85,041	$74	37%
Total 2020–2024	₩911,232	$788	₩234,429	$203	20%

* For the price level, the percentages at each year after patent expiration are the relative ratio compared with the price (=100%) at the time of patent expiration.

RESULTS

Figure 2. Cumulative budget savings according to the year of patent expiration during 2020–2024

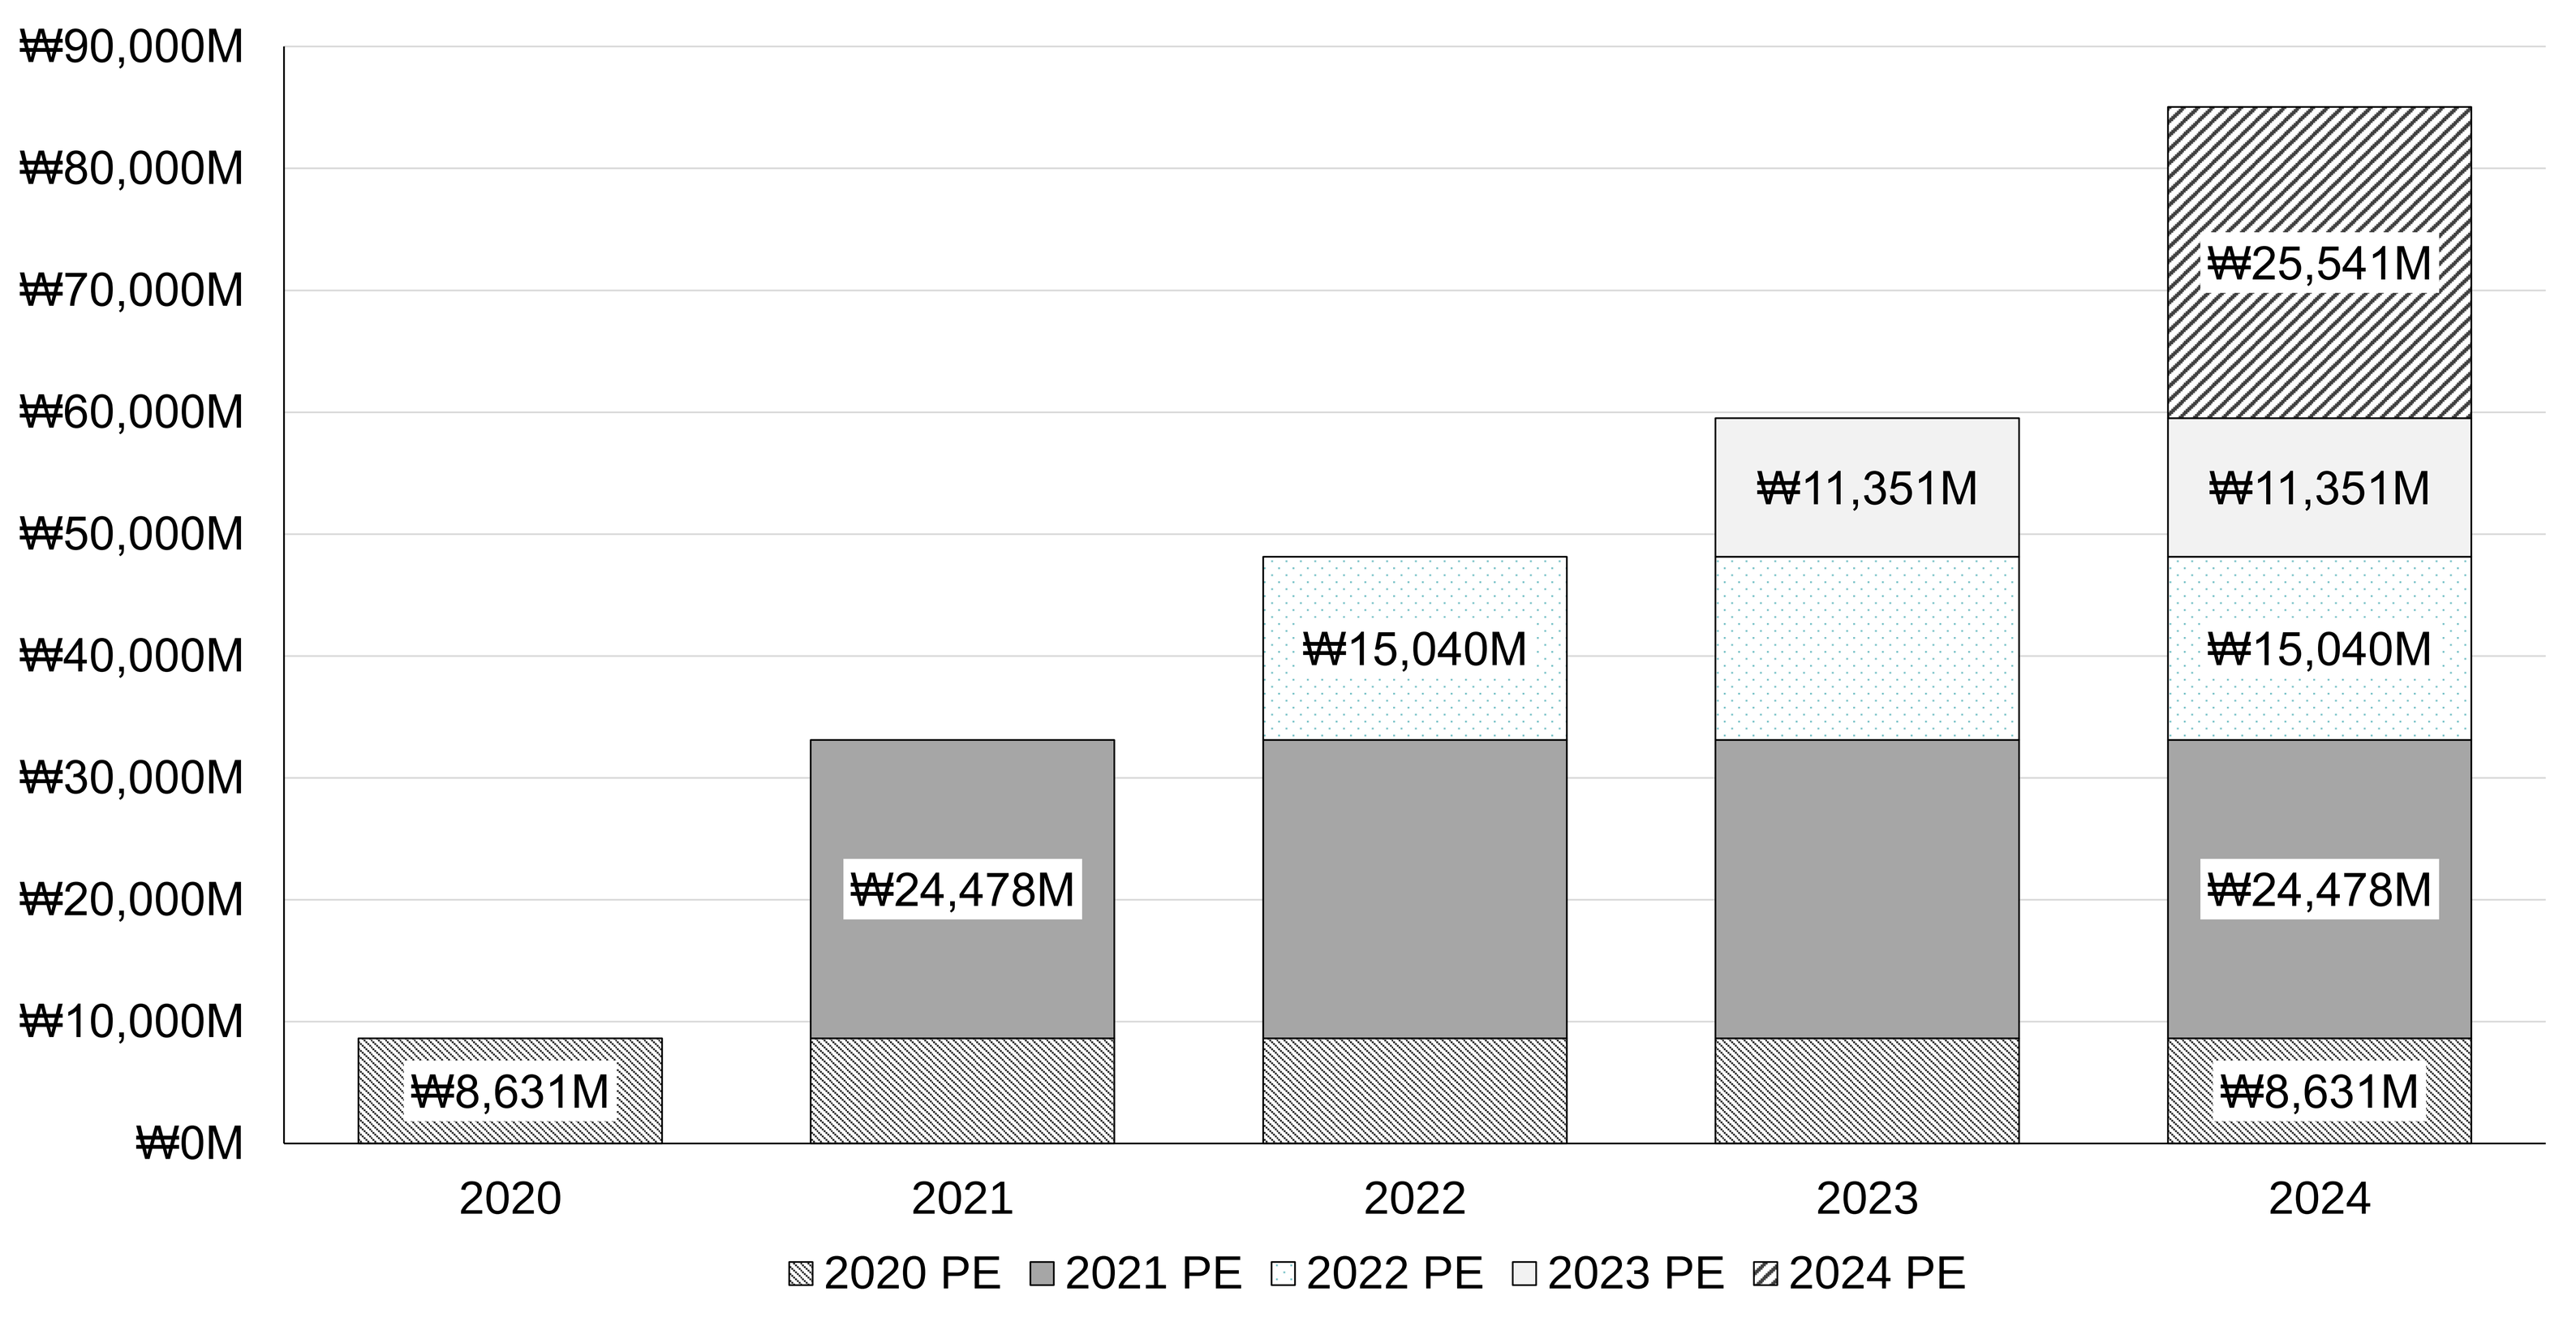
₩90,000M
₩80,000M
₩70,000M
₩60,000M
₩50,000M
₩40,000M
₩30,000M
₩20,000M
₩10,000M
₩0M
₩8,631M
₩24,478M
₩15,040M
₩11,351M
₩15,040M
₩24,478M
₩8,631M
₩25,541M
₩11,351M
2020
2021
2022
2023
2024
2020 PE
2021 PE
2022 PE
2023 PE
2024 PE

M: million Korean won (₩), PE: patent expiration

Figure 3. Sensitivity analyses for estimating the impact of input parameters on budget savings during 2020–2024

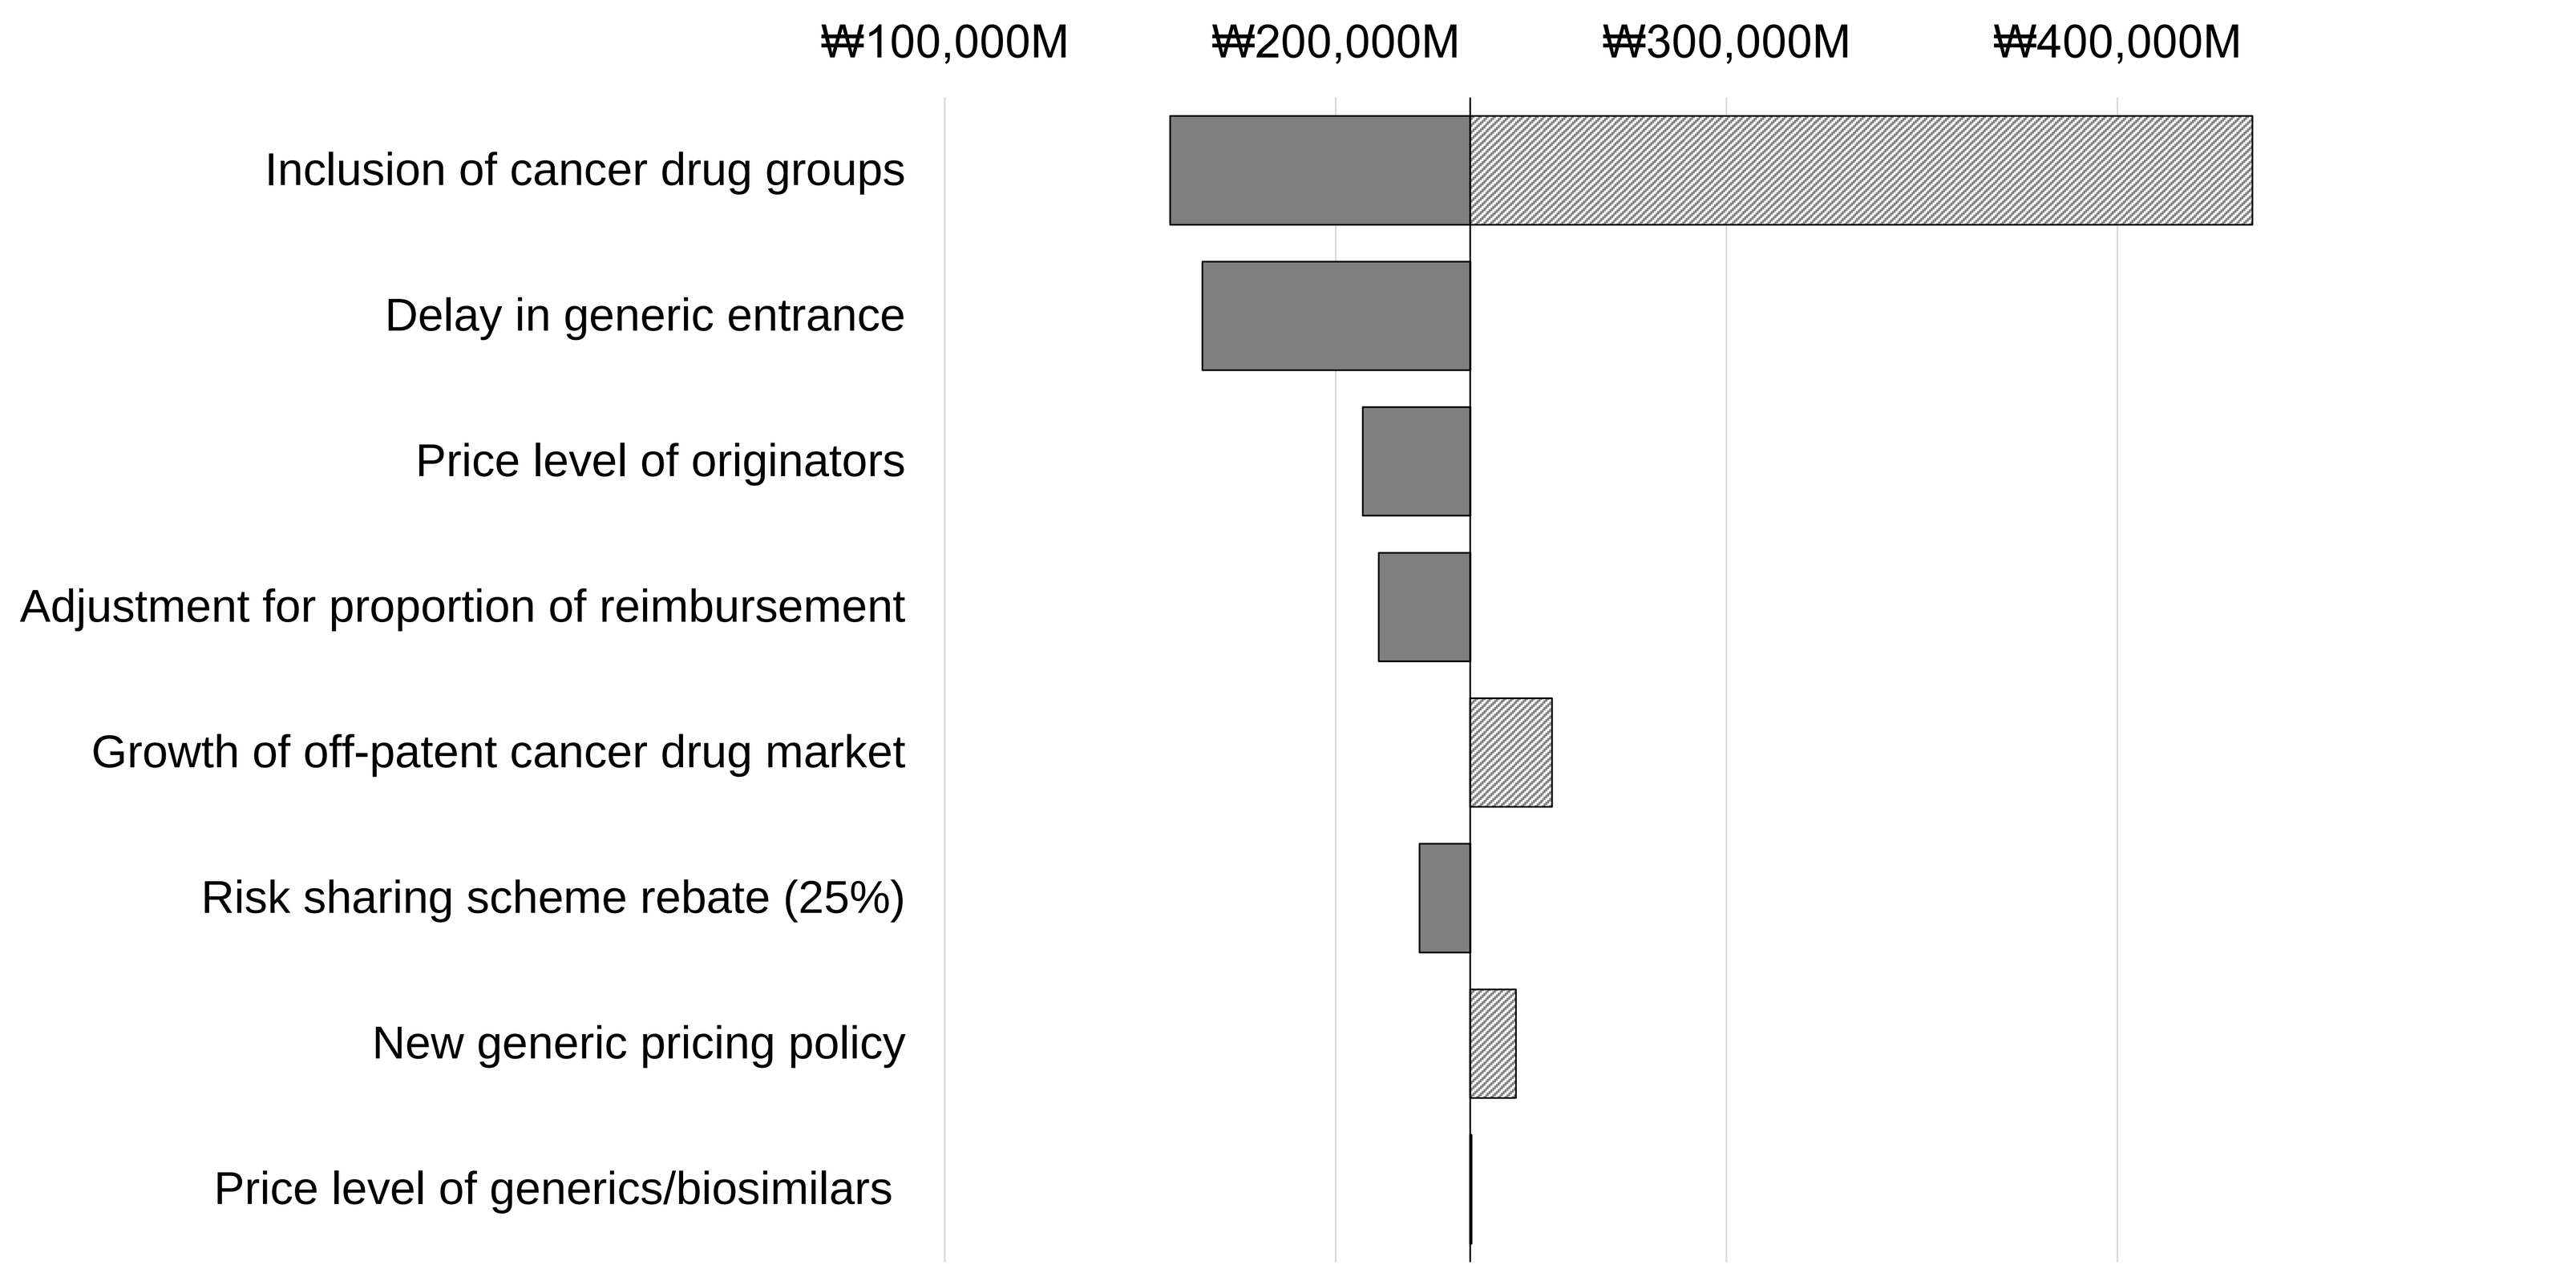
₩100,000M
₩200,000M
₩300,000M
₩400,000M
Inclusion of cancer drug groups
Delay in generic entrance
Price level of originators
Adjustment for proportion of reimbursement
Growth of off-patent cancer drug market
Risk sharing scheme rebate (25%)
New generic pricing policy
Price level of generics/biosimilars

M: million Korean won (₩), PE: patent expiration

Table 3. Total savings from patent expiration during 2020–2024 for various scenarios

Scenario	Budget savings			Changes from base case savings		
	Korea (₩): million	US ($): million	% of expenditures*	Korea (₩): million	US ($): million	(%)
Base case	₩234,429	$203	20%			
Based on group B cancer drugs	₩157,633	$136	14%	- ₩76,795	-$66	-33%
Based on group C cancer drugs	₩434,523	$376	38%	₩200,095	$173	+85%
Generic entrance: 6 months delay	₩165,904	$144	14%	- ₩68,524	-$59	-29%
Risk-sharing scheme rebate of 25%	₩221,465	$192	19%	-₩12,963	-$11	-6%
Adjustment for reimbursement	₩210,985	$183	18%	- ₩23,443	-$20	-10%
Growth of off-patent market	₩255,316	$221	22%	₩20,888	$18	+9%
Price level of originators	₩206,929	$179	18%	-₩27,499	-$24	-12%
Price levels of generics/biosimilars	₩234,771	$203	20%	₩343	$0	0%
New pricing policy (July 2020)	₩246,082	$213	21%	₩11,654	$10	+5%

* The proportion of budget savings was calculated by dividing budget savings for each scenario by the 5-year total expenditure for the base case (₩1,145,470 million).

CONCLUSIONS

- ❑ This study estimated that the patent expiration of originators will lead to total budget savings in cancer drug expenditures of ₩234,429 million ($203 million), ranging from ₩157,633 million ($136 million) to ₩434,523 million ($376 million) over 5 years.
- ❑ Budget savings (20% of the 5-year cancer drug expenditure) could be used to treat an additional of 94,000 cancer patients.
- ❑ The reallocation of these savings could improve the affordability of innovative and advanced cancer drugs, eventually improving the quality of life of patients with cancer.
- ❑ The actual savings would be higher if the savings were estimated based on various therapeutic classes of drugs that will be off-patent.
- ❑ Further studies are recommended to estimate the magnitude of budget impact for all therapeutic areas and to determine mechanisms of allocating limited resources more efficiently in Korea.

REFERENCES

1. Lindsley CW: New 2016 data and statistics for global pharmaceutical products and projections through 2017. ACS Chem Neurosci 2017;8(8):1635-1636
2. IQVIA Institute for Human Data Science. Global Oncology Trends 2018. 2018; https://www.iqvia.com/insights/the-iqvia-institute/reports/global-oncology-trends-2018. Accessed March 2, 2020.
3. Kenneally M, Walshe V: Pharmaceutical cost-containment policies and sustainability: recent Irish experience. Value Health 2012;15:389-393
4. Vondeling GT, Cao Q, Postma MJ, Rozenbaum MH: The impact of patent expiry on drug prices: A systematic literature review. Appl Health Econ Health Policy 2018;16:653-660

Funding: This research was partially funded by an unrestricted grant from MSD Korea Co. Ltd.