A comparison of economic outcomes between adherent and nonadherent patients treated with oral antidiabetic medications

Richard A. Hansen, PhD[1], Joel F. Farley, PhD[1], Marcus Droege, PhD[2], Matthew L. Maciejewski, PhD[3]

[1]University of North Carolina at Chapel Hill, UNC Eshelman School of Pharmacy, Division of Pharmaceutical Outcomes and Policy, Chapel Hill, NC; [2]Takeda Pharmaceuticals North America, Inc., Deerfield, IL;
[3]Durham Veterans Affairs Medical Center, Center for Health Services Research in Primary Care, Durham, NC, and Duke University Medical Center, Division of General Internal Medicine, Department of Medicine, Durham, NC

PDB10
ISPOR 2009

ABSTRACT

Objectives: Healthcare costs for adherent patients were compared with costs for nonadherent patients in a cohort with diabetes mellitus taking metformin, sulfonylureas, or pioglitazone.

Methods: Using both the commercial and Medicare supplemental MEDSTAT MarketScan® research databases, a retrospective cohort study identified 108,592 continuously insured patients 18 to 90 years of age with diabetes mellitus (ICD-9-CM 250.xx) using metformin, sulfonylureas, or pioglitazone monotherapy during 2003. Adherence was calculated in 2004 and 2005 using a medication possession ratio, and dichotomized at ≥80% to indicate adherence. Total healthcare costs included insurer payments and patient cost-sharing from medical (inpatient, outpatient) and prescription drug claims. Diabetes-specific costs included pharmacy claims for any diabetes drug and medical claims containing a primary diagnosis of diabetes or a diabetes complication. Multivariate analysis was conducted to adjust for potential confounders.

Results: During the initial year of follow-up, the proportion of patients adherent to treatment were 57%, 61%, and 59% for metformin, sulfonylureas, anad pioglitazone, respectively. Annual unadjusted total healthcare costs were consistently lower for adherent patients than for non-adherent patients ($1,470 less for metformin users, $3,734 less for sulfonylurea users, and $3,477 for pioglitazone users). In the multivariate models, total healthcare costs and diabetes specific healthcare costs respectively were $846 (95% CI $747 to $945) and $55 (95% CI $33 to $77) per year lower for adherent patients compared to nonadherent patients. The adjusted annual adherence-related cost difference was $336 for metformin (95% CI $216 to $456), $1,509 for sulfonylureas (95% CI $1,339 to $1,679), and $1,140 for pioglitazone (95% CI $793 to $1,486). Annual cost differences were larger among Medicare plans compared with commercial insurance plans ($2,220 lower vs $65 higher, respectively).

Conclusions: Adherence with oral diabetes medications results in overall healthcare cost-savings. Both sulfonylureas and pioglitazone were associated with larger total cost-savings than metformin.

OBJECTIVES

Benefits of oral antidiabetic medications are only realized when patients are adherent to treatment. Problems with oral antidiabetic medication adherence negatively affect clinical and economic outcomes, although little is known about whether the choice of oral medication affects the magnitude of the adherence-outcome relationship.
The objectives of this study were:

- To examine differences in medication adherence between patients taking metformin, sulfonylureas, or pioglitazone.
- To compare overall and diabetes-specific healthcare costs between adherent and non-adherent patients taking metformin, sulfonylureas, or pioglitazone, and to determine whether cost differences vary by medication.

METHODS

Study Design: A retrospective cohort study of patients 18 to 90 years of age with type 2 diabetes (ICD-9 250.xx) taking metformin, sulfonylureas, or pioglitazone was conducted.

Data Source: Data were derived from the commercial and Medicare supplemental files of the MEDSTAT MarketScan® Research Databases for the years 2003 – 2005.

Study Patients: Patients were assigned to one of three monotherapy cohorts based on 2003 medication use. For the economic analyses, patients meeting eligibility criteria during 2003 were followed during 2004 and 2005 using an intention-to-treat approach; patients were followed regardless of whether they continued to use an eligible drug.

Included N=200,745
- Continuous insurance eligiblity 2003–2005
- Aged 18-90 years during 2003.
- ≥2 outpatient or ≥1 inpatient diagnosis code for diabetes (ICD-9 250.xx) during 2003
- ≥2 claims for eligible OHA during 2003.

Excluded N=200,745
- Combination products and polytherapy.

N=108,592
Monotherapy

Metformin (N=52,156)

Sulfonylurea (N=44,916)

Pioglitazone (N=11,520)

Key Variables:

Dependent: Costs

Healthcare costs were estimated for claims for inpatient care, outpatient care, and prescription drugs. All expenditures were adjusted to 2005 dollars using the medical component of the Consumer Price Index.[1] All-cause total healthcare expenditures were estimated for each patient in each year by summing payments from all sources with beneficiary cost-sharing amounts. Diabetes-specific costs were estimated by claims for which an ICD-9 code for diabetes or a complication of diabetes appeared in the primary diagnosis field.[2]

Independent: Adherence

Defined by the medication possession ratio (MPR), calculated as the ratio of the total days supply of a medication divided by the number of days in the evaluation period.[3] Adherence was dichotomized by classifying individuals as adherent if their MPR was ≥80%, and non-adherent if their MPR was <80%.

Covariates
Age, sex, region, insurance type, insurance origin, comorbidity,[4] prescription cost-sharing amount, and medication switching were controlled in multivariate models.

Analysis
A generalized linear model (GLM) with a gamma distribution and square root link was used to model 2004 and 2005 all-cause total healthcare costs on adherence for the three drug cohorts in a pooled sample. The key explanatory variables of interest were an indicator for adherence, three indicators for the three drug cohorts, and interaction terms of adherence and each drug cohort. To compare adjusted costs between adherent and non-adherent users in each drug cohort, predicted costs for every person in the pooled sample were estimated. In a sensitivity analysis, we estimated all-cause total costs and diabetes-specific total costs separately for the commercial plan sample and Medicare supplemental insurance sample. Sensitivity analyses also estimated inpatient, outpatient, and prescription costs. One-part models were estimated for outpatient and prescription drug costs, but two-part models were estimated for inpatient costs to account for the large proportion of patients receiving no inpatient care.

RESULTS

Population Characteristics

The majority of patients were older than 45 years of age (mean=63 years). The proportion of patients residing in the north-central, south, and west regions of the United States were generally similar (approximately 30% for each); fewer patients from the north-east region contributed data. The majority of patients had either fee-for-service insurance (ie, major medical), or were covered by a health maintenance organization (HMO) or preferred provider organization (PPO) plan.

Comparing patient characteristics among the sulfonylurea cohort and pioglitazone cohort with the metformin cohort, the metformin cohort represented a greater proportion of female patients and was generally healthier (ie, smaller comorbidity index) than the other cohorts; the sulfonylurea cohort was generally older; and the pioglitazone cohort mostly comprised the north-central and south regions, represented a greater proportion of patients with major medical or PPO insurance, and patients paid higher drug coinsurance or co-payments for this drug.

Baseline characteristics by index drug type

Characteristic[a]	Metformin N=52,156	Sulfonylurea N=44,916	Pioglitazone N=11,520
Age, Years (%)			
18 to 34	2.3	0.9	1.4
35 to 44	8.6	4.2	5.9
45 to 54	25.1	14.8	22.3
55 to 64	26.6	19.9	29.4
≥65	37.3	60.3	41.0
Female (%)	52.8	47.2	46.1
Regional Residence, (%)			
North-East	5.5	5.5	6.2
North-Central	29.2	30.8	43.2
South	33.1	31.7	38.0
West	31.8	31.7	12.2
Unknown Region	0.4	0.3	0.4
Insurance Type (%)			
Major Medical	26.2	34.3	44.1
HMO	33.1	36.4	10.3
POS	6.4	4.4	7.3
PPO	32.1	23.6	36.2
Unknown/Other	0.2	0.2	0.2
Insurance Type (%)			
Disability or COBRA	0.4	0.3	0.5
Employer (Full- and Part-Time)	33.8	20.5	32.1
Early Retiree	11.4	8.6	15.7
Medicare Eligible Retiree	16.5	26.2	31.1
Other Retiree	5.4	6.2	6.8
Surviving Spouse	2.6	4.4	4.4
Unknown	29.9	33.7	9.3
Comorbidity,b Mean (SD)	1.45 (1.02)	1.78 (1.34)	1.69 (1.27)
Copayment,c Mean (SD)	$13.3 ($12.4)	$12.4 ($9.8)	$21.9 ($24.8)

POS=point of service; COBRA=Consolidated Omnibus Budget Reconciliation Act.
[a]Based on sample selection criteria applied during 2003.
[b]Based on Deyo derivation of the Charlson comorbidity index.[4]
[c]Reflects mean (SD) cost-sharing amount per prescription fill.

Adherence profile by drug and year

Treatment	2004 MPR Mean (SD)	2004 Adherent (%)	2005 MPR Mean (SD)	2005 Adherent (%)
Metformin (n=52,156)	**70.9 (33.6)**	**56.7**	**66.7 (36.4)**	**53.0**
Sulfonylurea (n=44,916)	**73.8 (32.8)**	**61.0**	**67.5 (37.2)**	**55.5**
Chlorpropamide (n=247)	73.9 (35.0)	63.6	59.9 (40.7)	48.6
Glimepiride (n=8,609)	71.5 (34.3)	58.7	63.9 (39.4)	52.7
Glipizide (n=18,185)	74.4 (33.3)	62.5	66.5 (38.6)	55.9
Glyburide (n=16,056)	72.9 (32.9)	59.5	65.0 (37.8)	65.0
Tolazamide (n=485)	76.9 (31.9)	66.8	65.1 (38.7)	53.0
Tolbutamide (n=60)	79.4 (31.1)	71.7	71.7 (36.7)	60.0
Pioglitazone (n=11,520)	**70.2 (36.3)**	**59.4**	**61.3 (41.3)**	**52.5**

Adjusted annual costs and cost differences by adherence

Total Costs (US Dollars)	Mean Annual Cost[a] Adherent	Mean Annual Cost[a] Non-Adherent	Cost Difference for Non-Adherent Compared with Adherent: Difference	Cost Difference for Non-Adherent Compared with Adherent: 95% Confidence Interval
All-Cause				
Overall[b] (n=108,592)	$12,412	$13,258	$846	$747 to $945
Metformin (n=52,156)	$11,152	$11,488	$336	$216 to $456
Sulfonylureas (n=44,916)	$12,759	$14,268	$1,509	$1,339 to $1,679
Pioglitazone (n=11,520)	$16,596	$17,736	$1,140	$793 to $1,486
Primary Diabetes or Complication				
Overall[b] (n=108,592)	$2,230	$2,284	$55	$33 to $77
Metformin (n=52,156)	$1,870	$1,864	($6)	($31) to $20
Sulfonylureas (n=44,916)	$2,114	$2,385	$271	$235 to $307
Pioglitazone (n=11,520)	$4,295	$3,862	($433)	($516) to ($350)

[a]Predicted values from 2004 and 2005 are inflation-adjusted and derived from a general linear model of the pooled population controlling for medication changes, sex, age, region, insurance type, insurance origin, comorbidity, and cost-sharing.

[b]Overall represents a pooled estimate for metformin, sulfonylureas, and pioglitazone.

Adjusted annual cost difference between nonadherent patients and adherent patients by drug, population, and cost type for 2004 and 2005

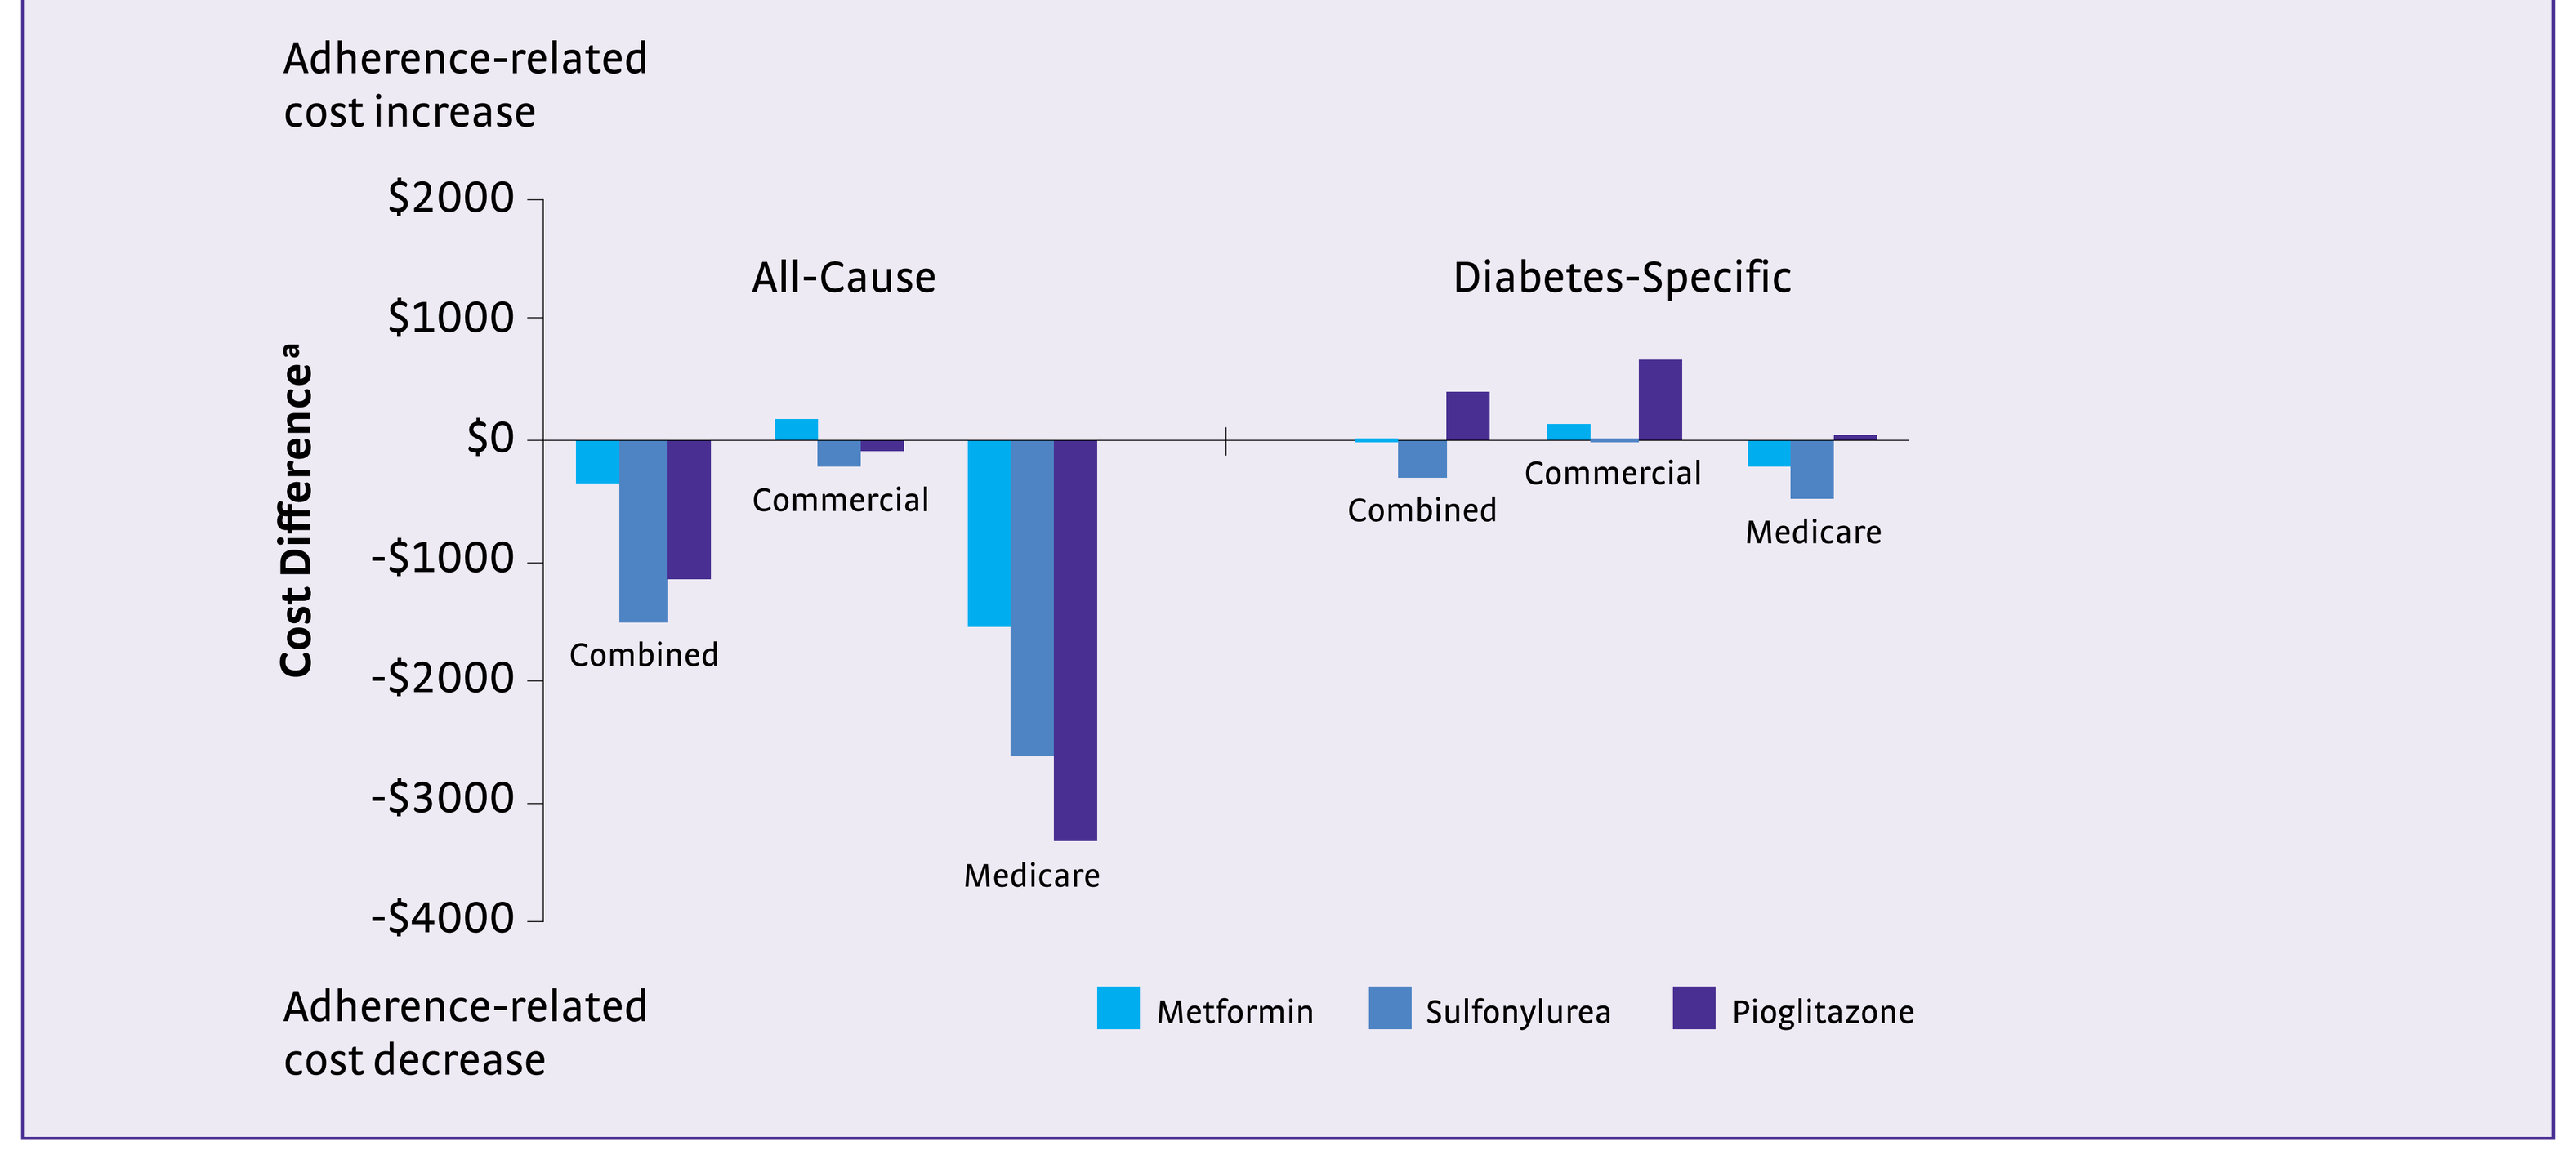

Adherence-related cost increase
$2000
$1000
$0
-$1000
-$2000
-$3000
-$4000
Cost Difference[a]
All-Cause
Diabetes-Specific
Combined
Commercial
Medicare
Adherence-related cost decrease
Metformin
Sulfonylurea
Pioglitazone

[a]Predicted values from 2004 and 2005 are inflation- adjusted and derived from a general linear model of the pooled monotherapy population controlling for medication changes, sex, age, region, insurance type, insurance origin, comorbidity, and cost-sharing.

Adjusted all-cause annual costs and cost differences by adherence and cost source

Total Costs (US Dollars)	Mean Annual Cost[a] Adherent	Mean Annual Cost[a] Non-Adherent	Cost Difference for Non-Adherent Compared with Adherent: Difference[b]	95% Confidence Interval	
Inpatient[c] (n=108,592)	$2,116	$3,904	$1,789*	$1,751 to	$1,826
Outpatient (n=108,592)	$5,411	$6,400	$988*	$938 to	$1,039
Prescription (n=108,592)	$4,331	$3,503	($828)*	($839) to	($817)

*P<0.05.

[a]Predicted values from 2004 and 2005 are inflation-adjusted and derived from a general linear model of the pooled population controlling for medication changes, sex, age, region, insurance type, insurance origin, comorbidity, and cost-sharing.

[b]Sum of adjusted component costs does not equal the overall adjusted cost difference because of covariate adjustment and distributional assumptions in each component model.

[c]Inpatient predictions were estimated using two-part models. Part 1 estimates the probability of receiving any inpatient care using logistic regression. Part 2 estimates the cost of care among those receiving care using GLM.

CONCLUSIONS

This study further substantiates the importance of oral antidiabetic medication adherence, illustrating the overall net cost-savings for adherent patients compared with nonadherent patients.

- Mean annual adherence was similar among agents during the first year of follow-up (ranging from 70% to 74%), and all-cause annual costs were $846 per year less for adherent patients compared with nonadherent patients.
- Sulfonylureas and pioglitazone were associated with larger adherence-related cost-savings than metformin.
- The Medicare sample accounted for the majority of adherence related cost-savings ($2,220/year).
- Cost-savings were primarily derived from reduced inpatient and outpatient care.
- Limitations include unobserved confounding, use of a prevalent user design, implications of intention-to-treat analysis, and limitations inherent in using pharmacy claims data to measure adherence.

REFERENCES

1. U.S. Department of Labor. Consumer Price Index Summary. Available at www.bls.gov/cpi. Accessed July 30, 2008.
2. Bethel MA, Sloan FA, Belsky D, et al. Longitudinal incidence and prevalence of adverse outcomes of diabetes mellitus in elderly patients. *Arch Intern Med* 2007;167:921-927.
3. Andrade SE, Kahler KH, Frech F, et al. Methods for evaluation of medication adherence and persistence using automated databases. *Pharmacoepidemiol Drug Saf* 2006;15:565-574.
4. Deyo RA, Cherkin DC, Ciol MA. Adapting a clinical comorbidity index for use with ICD-9-CM administrative databases. *J Clin Epidemiol* 1992;45:613-619.